C-FOS Expression in the Periaqueductal Gray Varies Relative to the Method of Conditioned Taste Aversion Extinction Employed

G. Andrew Mickley, Gina N. Wilson, Jennifer Remus, Linnet Ramos, Kyle Ketchesin, Orion Biesan, Joseph Luchsinger and Suzanna Prodan

The Neuroscience Program and Department of Psychology, Baldwin-Wallace College, Berea, OH 44017 USA

Abstract

A conditioned taste aversion (CTA) is acquired when an animal consumes a novel taste (CS) and then experiences the symptoms of poisoning (US). Following CTA training, animals will avoid the taste that was previously associated with malaise. This defensive reaction to a learned fear can be extinguished by repeated exposure to the CS alone. However, following a latency period in which the CS is not presented, the CTA will spontaneously recover (SR). Thomas *et al.* (2005) have used an explicitly unpaired (EU) extinction procedure that disassociates a light CS and footshock to thwart renewal of a conditioned emotional response. Here we applied similar procedures to the CTA paradigm and also evaluated the ability of EU extinction procedures to affect behavioral indicators of SR and *c-fos* expression. Fluid-deprived Sprague-Dawley rats acquired a CTA [3 pairings of 0.3% oral saccharin (SAC; the CS) and 81mg/kg i.p. lithium chloride (LiCl; the US)] followed by extinction trials consisting of multiple exposures to either, (a) CS-only, or (b) CS and US on alternate days (EU extinction). Both extinction procedures resulted in ≥90% reacceptance of SAC and were followed by a 30-day latency period of water drinking. Rats were then tested for SR with a final exposure to SAC before sacrificing. Brain *c-fos* protein expression was evaluated via immunohistochemistry. Rats in the CS-only group exhibited significant suppression of SAC drinking during their SR test compared to their consumption at the end of extinction. However, animals in the EU extinction group did not show such SR of the CTA and drank significantly more than the CS-only rats. The brains of EU-extinguished rats and CS-only extinguished rats did not differ in the number of *c-fos*-labeled neurons in gustatory neocortex, medial prefrontal cortex, basolateral amygdala or the central nucleus of the amygdala. However some small, but reliable, differences were detected in Periaqueductal gray (PAG) – especially in the dorsolateral region. Thus, behavioral differences in SR between the EU and CS-only extinction animals were not represented by corresponding changes in the neural activity of several brain nuclei classically associated with extinction learning. However a detailed analysis of PAG *c-fos* expression provides hints about some of the physiological changes evoked by these 2 extinction paradigms. The findings are clinically relevant as we seek the development of treatments for deficits in fear extinction (e.g. PTSD, phobias).

Introduction

- Animals will avoid a tastant that has previously been associated with malaise (Garcia et al., 1955).
- Like other defensive reactions to conditioned fears (e.g., PTSD and phobias), the conditioned taste aversion (CTA) is resistant to extinction (EXT) and prone to spontaneous recovery (SR) (Parker, 2003).
- However, by explicitly unpairing the CS and the US during the extinction process (EU-EXT), both the time to reach asymptotic extinction and spontaneous recovery of the CTA may be reduced significantly when compared to animals that received CS-only (CSO) (Mickley et al., 2009).
- We have been using *c-fos* protein immunohistochemistry as an indicator of neural activity in order to investigate the neural substrate of CTA extinction and SR (Lamprecht and Dudai, 1996).
- Depending on the time during the extinction process sampled, medial Prefrontal Cortex (mPFC), Basolateral Amygdala (BLA), and Gustatory Neocortex (GNC) are active during CTA extinction (Mickley et al., 2004; 2005). Further, mPFC and GNC adjust their activity during spontaneous recovery of the CTA (Mickley et al, 2007).
- The current study sought to characterize differences in patterns of *c-fos* protein expression in rats that underwent CSO extinction vs. EU-EXT.
- Here, we extended our analysis to the midbrain **Periaqueductal Gray (PAG)** since it is functionally connected to the mPFC, amygdala (AMY) and other structures that have been implicated in the modulation of emotional responses (Pare et al., 2004; Price, 2005; Peters et al., 2009; Ulrich-Lai and Herman, 2009; Floyd et al., 2000) and extinction of conditioned fears (McNally et al., 2004; 2005).

Methods

Subjects: Male, Sprague-Dawley rats

Conditioning

- Animals were habituated to a 23h water deprivation schedule for 2 days prior to the start of the experiment.
- On days 1, 3, and 5 of CTA conditioning, animals were presented with a 0.3 % saccharin solution (SAC) for 30 min. During a 15 min latency animals were injected with lithium chloride (LiCl; 81mg/kg, i.p.) and subsequently presented with tap water for 30 min to prevent dehydration.
- Days 2, 4, and 6 served as rest days during which the animals received two 30 min presentations of water separated by 15 min.

Extinction

- **CS-Only Extinction:** Animals received 30 min SAC exposure and 15 min later were presented with water for 30 min every-other day until reaching asymptotic extinction (90% baseline SAC consumption levels; Nolan et al., 1997).
- **Explicitly Unpaired Extinction (EU-EXT):** Animals received 30 min SAC exposure followed 15 min later by 30 min water presentation every-other day of the extinction phase until reaching asymptotic extinction. On alternate days these animals received two 30 min presentations of water separated by a 15min latency during which LiCl (81mg/kg, i.p.) was administered.

Spontaneous Recovery (SR)

- After reaching asymptotic extinction, animals were, each day, given two 30 min presentations of water for 29 days.
- On day 30 following asymptotic extinction, animals were allowed to drink SAC for 30 min.

Histology

- Rats were transcardially perfused 90 min following their final SAC exposure.
- Brains were harvested and sliced at 40µm. Slices were collected and assayed for *c-fos* protein immunoreactivity, mounted and counterstained with neutral red. Cells staining positive for *c-fos* protein (Hsu et al., 1981) were counted. Only round, dark, uniformly stained cells were included. The observer (GAM) was blind to the experimental condition of the rats.
- Sections were viewed using light microscopy and Axiovision™ software.
- The PAG was located using standard demarcations from The Rat Brain in Stereotaxic Coordinates (Paxinos & Watson, 1998) between -7.2 and -8.2mm posterior to bregma.
- The PAG analysis was divided into the 4 recognized major longitudinal columns: Dorsomedial PAG (dmPAG); Dorsolateral PAG (dlPAG); Lateral PAG (lPAG) and Ventrolateral PAG (vlPAG).
- An α level of 0.05 was used to determine significance.

Table 1: Group Nomenclature and Treatments

	Group Designation	Conditioning Day 1	Day 2	Day 3	Day 4	Day 5	Day 6	Extinction[3]	30-day SR test	Number of rats per group[4]
CTA	CS-Only Extinction (CSO)	SAC[1]+LiCl[2]	Water	SAC+LiCl	Water	SAC+LiCl	Water	SAC	SAC	12/6/6
CTA	Explicitly Unpaired Extinction (EU-EXT)	SAC+LiCl	Water	SAC+LiCl	Water	SAC+LiCl	Water	SAC+LiCl	SAC	11/ 5/6
CTA	No Extinction (NE)	SAC+LiCl	Water	SAC+LiCl	Water	SAC+LiCl	Water	Water	N/A	16/9/7

[1]SAC = 30 min exposure to the 0.3%SAC solution

[2]LiCl = Injection of lithium chloride (81mg/kg, i.p.)

[3]Extinction = EU Extinction refers to the 2 different treatments received on alternate days throughout the extinction phase; SAC was administered every-other day to animals designated as CS-Only Extinction.

[4]The first number indicates the total number of rats in each group. The second number represents the number of rats that were sacrificed following asymptotic extinction. The third number represents the number of rats sacrificed following the spontaneous recovery test.

Results

Figure 1. CTA Acquisition: At the end of acquisition, rats from all 3 treatment groups (CSO, EU and NE groups; combined here) exhibited equally profound Conditioned Taste Aversions:

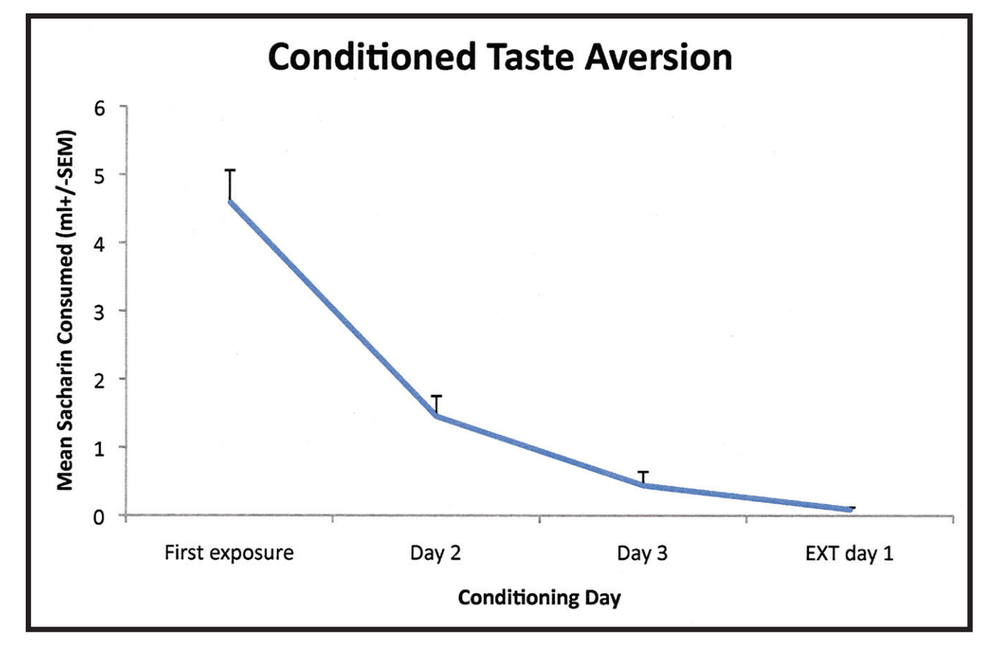
Conditioned Taste Aversion
Mean Sacharin Consumed (ml+/-SEM)
First exposure
Day 2
Day 3
EXT day 1
Conditioning Day

Figure 2. CTA Extinction: Rats exposed to the EU-EXT method extinguished the CTA rapidly and did not exhibit Spontaneous Recovery (SR) as compared to rats that experienced the CSO-EXT procedure.

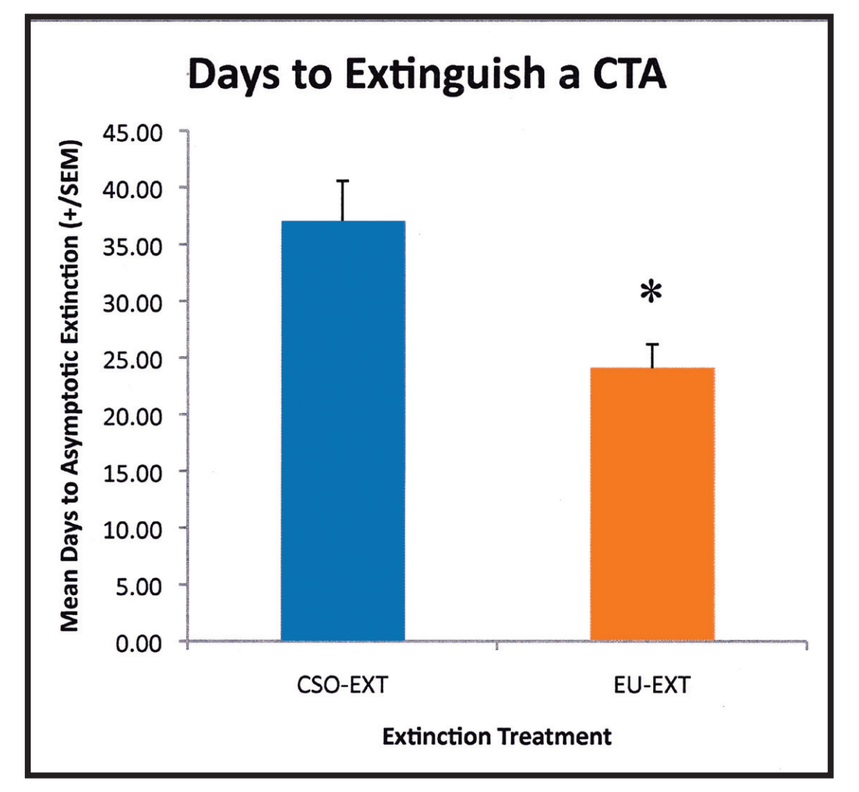
Days to Extinguish a CTA
Mean Days to Asymptotic Extinction (+/SEM)
CSO-EXT
EU-EXT
Extinction Treatment

The explicitly unpaired extinction group took significantly fewer days to extinguish the learned fear than the CS-only extinction group. * = Significantly different from the CS-Only Extinction Group: [t (21) = 3.00; p = 0.007]

Figure 3. SAC Consumption on the Day of Asymptotic Extinction or Spontaneous Recovery Test:

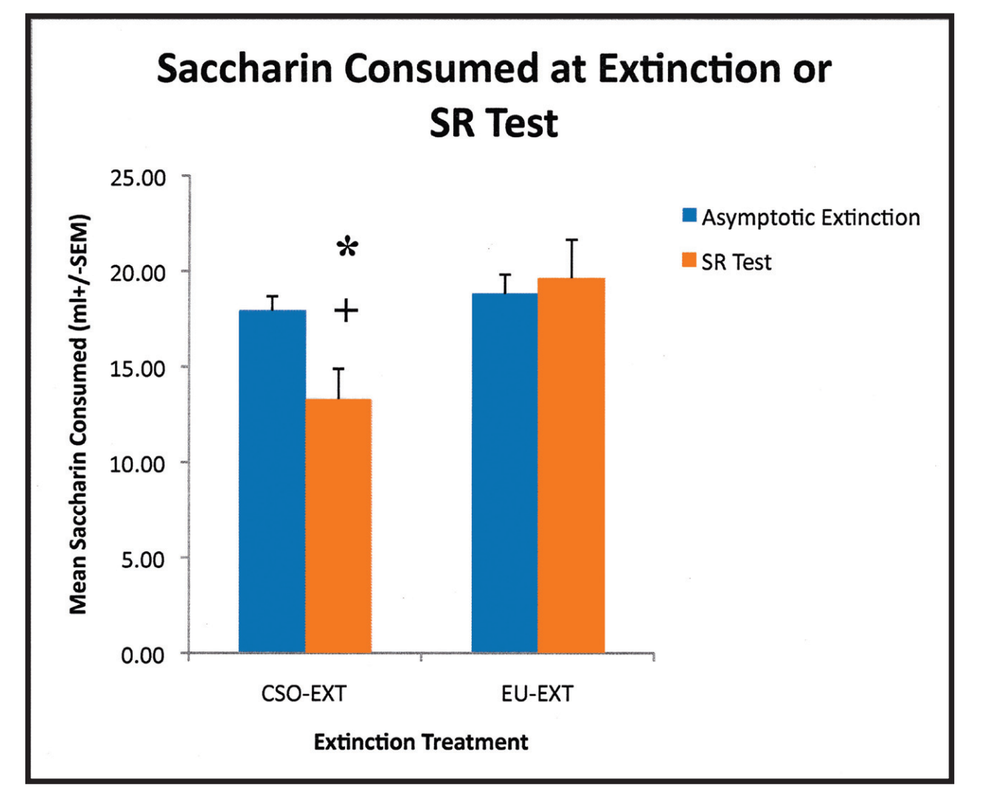
Saccharin Consumed at Extinction or SR Test
Asymptotic Extinction
SR Test
Mean Saccharin Consumed (ml+/-SEM)
CSO-EXT
EU-EXT
Extinction Treatment

The explicitly unpaired extinction group (EU-EXT) drank about the same amount of SAC on the day of extinction as they did on the 30-Day SR test day. However, the CS-only (CSO-EXT) extinction group drank significantly more SAC on the day of extinction than on the day of the SR test [t (5) = 2.72; p = 0.042; *]. Likewise, SAC drinking at the SR test was significantly less in the CS-only rats than in the EU-EXT rats [t (9) = 2.47; p = 0.036; +]. This indicates that the CS-only extinction animals had a SR of the CTA, but the EU-EXT group did not.

Figure 4. Individual Animal SAC Consumption Curves throughout Extinction: The extinction curves of the slowest animal to extinguish and fastest animal to extinguish from each of the two main experimental groups (EU-EXT = circles; CS-Only Extinction = inverted triangles).

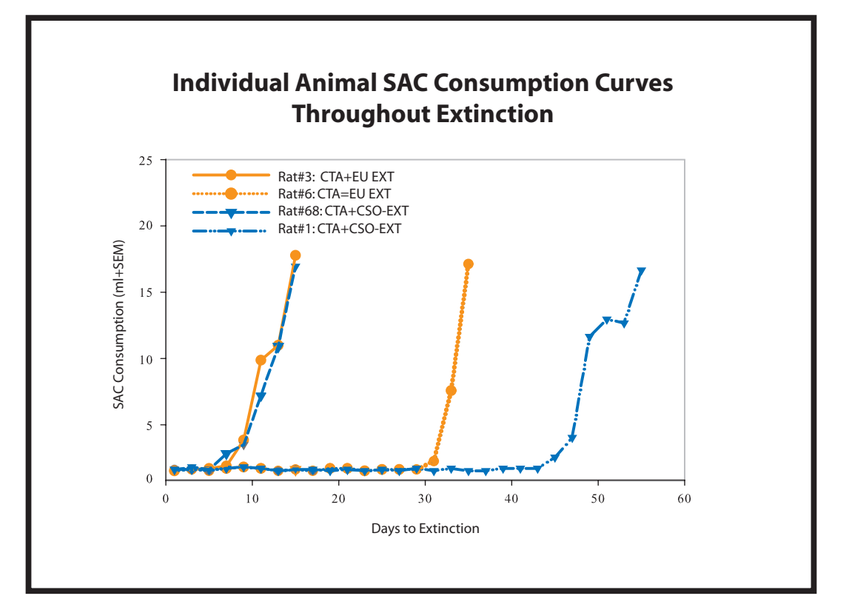
Individual Animal SAC Consumption Curves Throughout Extinction
Days to Extinction

Rats in the EU-EXT group took a smaller range of days to extinguish than the CS-Only Extinction group.

Figure 10. Following the SR test, rats that went through EU extinction contains proportionally more *c-fos*-labeled cells in the dlPAG than in other longitudinal columns of this midbrain structure.

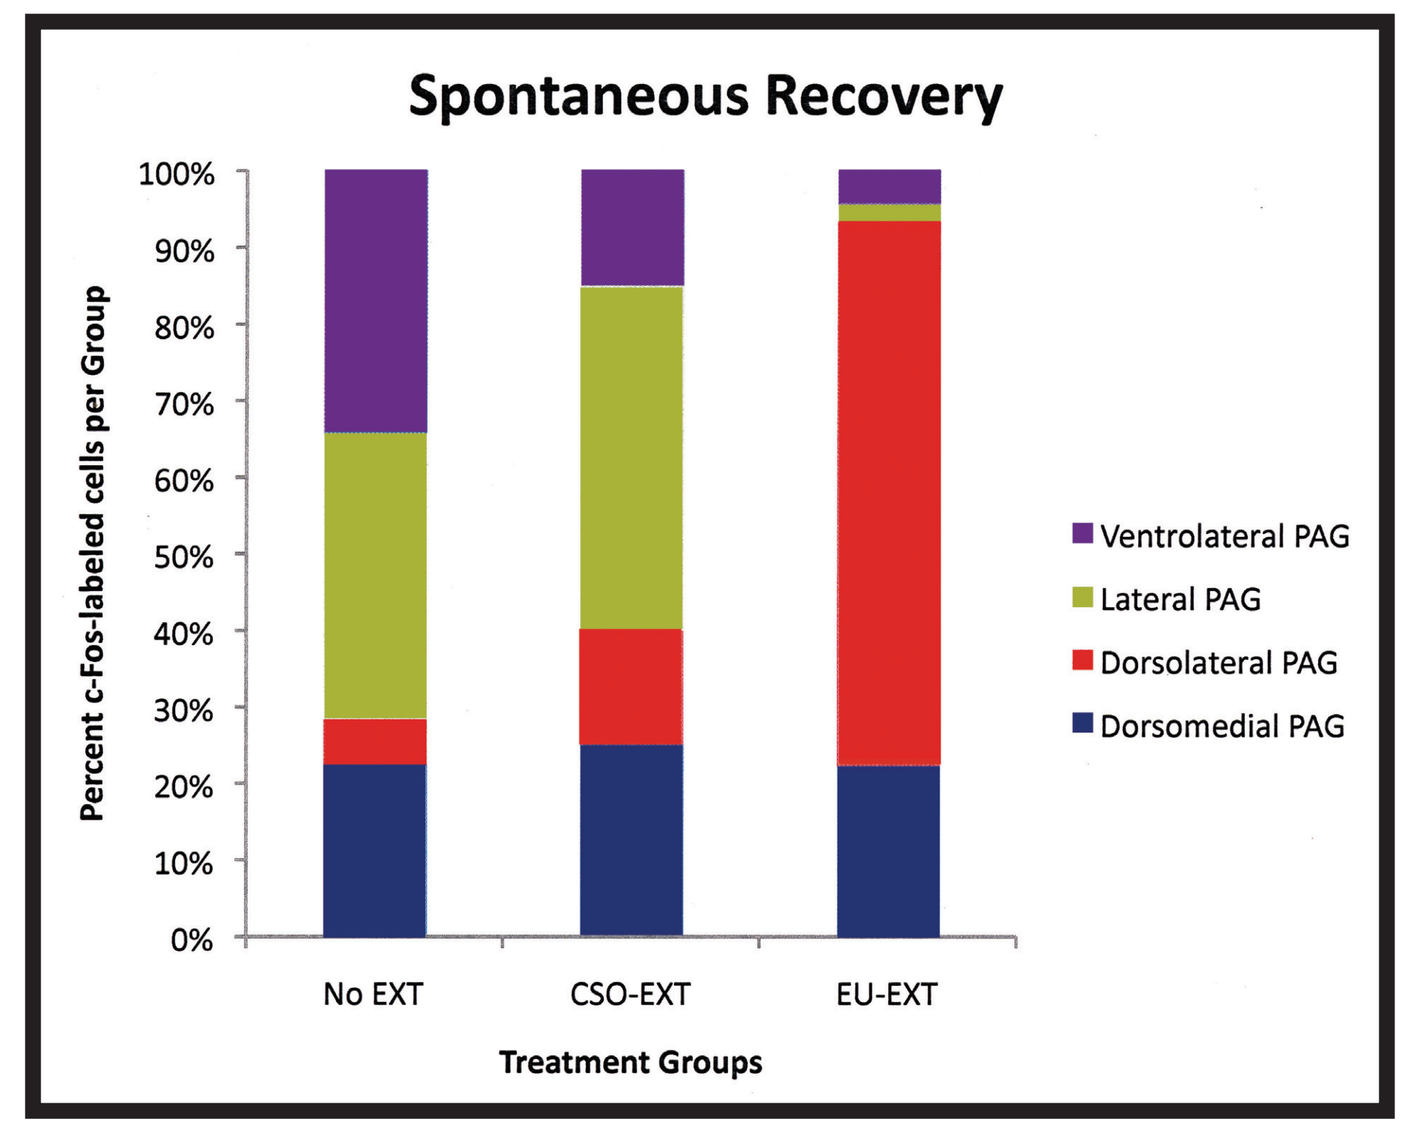
Spontaneous Recovery
Percent c-Fos-labeled cells per Group
Ventrolateral PAG
Lateral PAG
Dorsolateral PAG
Dorsomedial PAG
No EXT
CSO-EXT
EU-EXT
Treatment Groups

Figure 11. c-Fos expression in the dlPAG following SR test. After the SR test, rats that underwent the EU-EXT procedure expressed more *c-fos*-labeled neurons in the dorsolateral PAG than CS-Only extinction control animals.

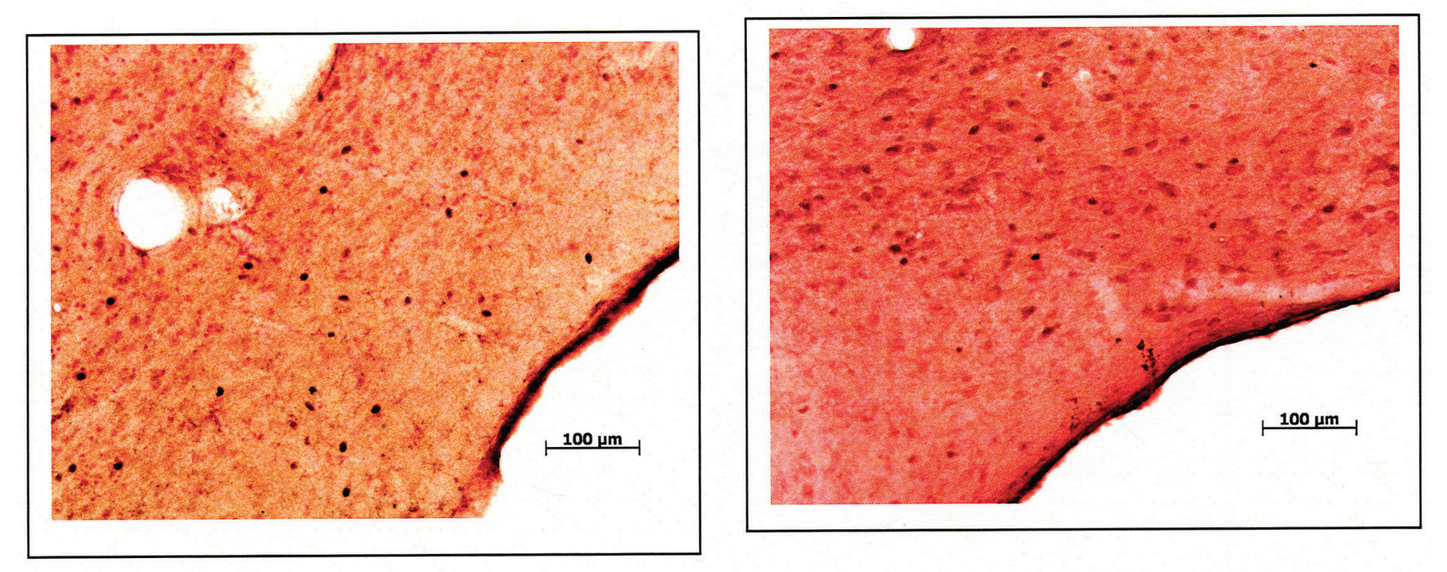

EU-EXT CS-Only EXT

Results

C-fos **Immunohistochemistry:**

- **The PAG from EU-EXT rats exhibited a small, but reliable, increase in *c-fos* expression as compared to the PAG of CS-Only animals following the SR test.**
- **As compared to rats that maintained their CTA (NE group), EU-EXT or CSO-EXT animals, having achieved asymptotic extinction, uniformly suppressed *c-fos* expression in all 4 longitudinal columns of the PAG.**
- **Rats that underwent the CSO or EU extinction procedures did not differ in the number of cells exhibiting *c-fos* expression in the PAG when they reached asymptotic EXT.**
- **Neurons in the dlPAG expressed more *c-fos* after the SR test if the rats experienced the EU-EXT procedure instead of the CSO-EXT procedure.**
- **No significant differences in *c-fos* expression in mPFC (prelimbic or infralimbic), GNC, or Amygdala (basolateral or central nuclei) were observed between EU-EXT and CS-Only Extinction brains following the SR test.**

Figures 5- 9. Number of *c-fos*-labeled cells in the entire PAG or its longitudinal columns.
* = Statistically significant difference between *c-fos* labeling at asymptotic extinction vs. SR. The horizontal bars represent significant differences between particular treatment groups. ANOVA [Extinction Treatment (CSO or EU) X Test day (Asymptotic EXT or SR)] and Bonferroni *post hoc* comparisons. α = 0.05.

Figure 5.

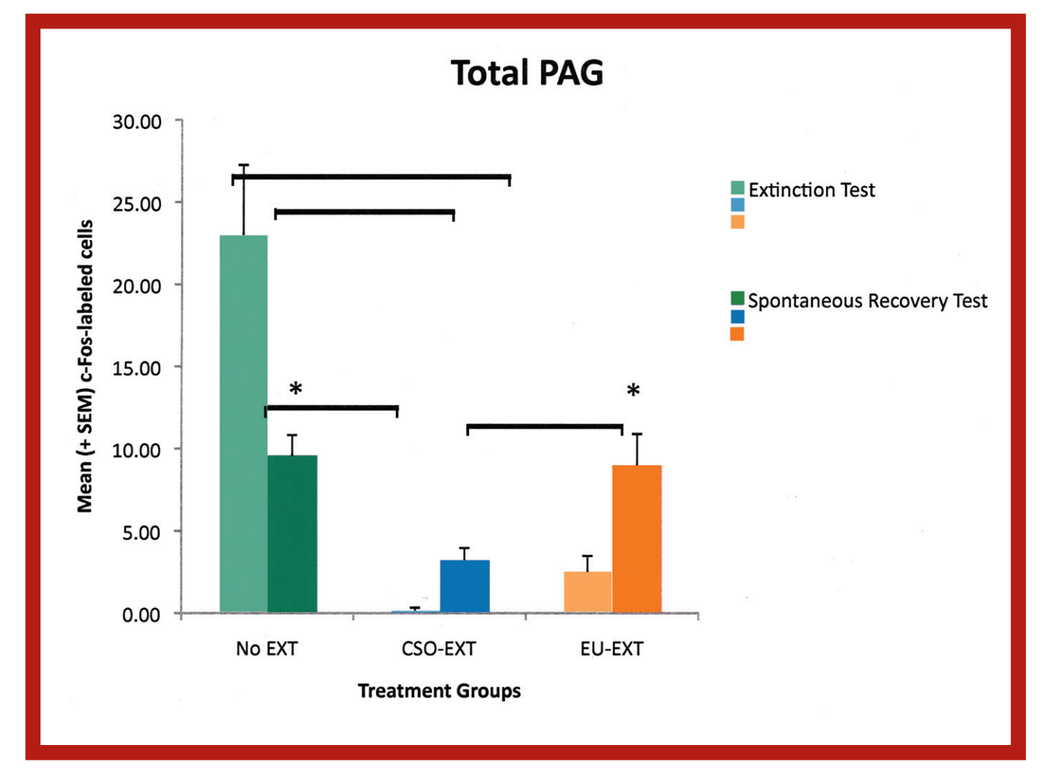
Total PAG
Extinction Test
Spontaneous Recovery Test
Mean (+ SEM) c-Fos-labeled cells
No EXT
CSO-EXT
EU-EXT
Treatment Groups

Figure 6:

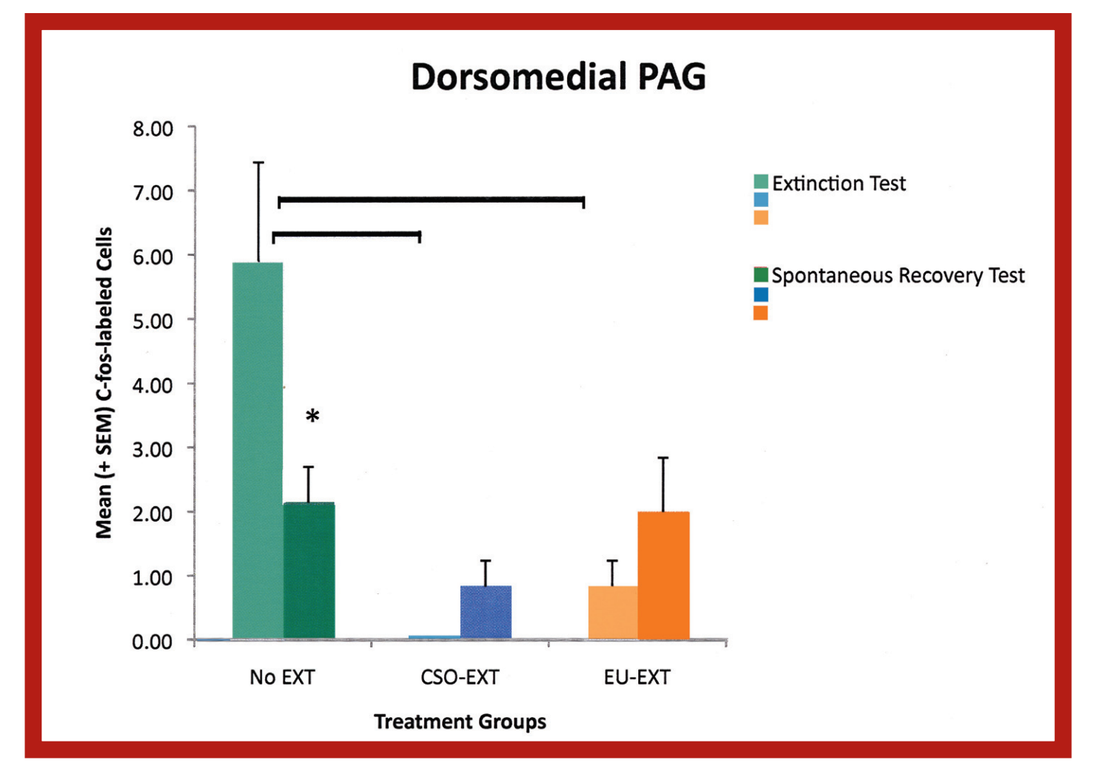
Dorsomedial PAG
Extinction Test
Spontaneous Recovery Test
Mean (+ SEM) C-fos-labeled Cells
No EXT
CSO-EXT
EU-EXT
Treatment Groups

Figure 7:

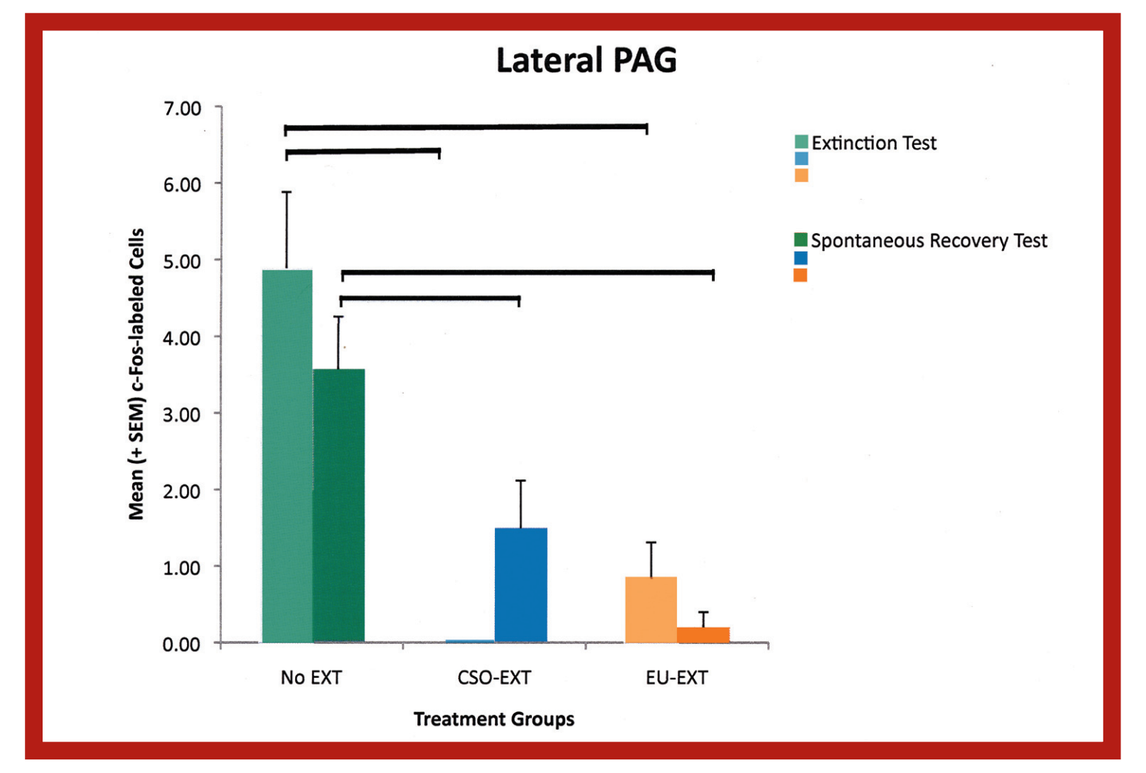
Lateral PAG
Extinction Test
Spontaneous Recovery Test
Mean (+ SEM) c-Fos-labeled Cells
No EXT
CSO-EXT
EU-EXT
Treatment Groups

Figure 8:

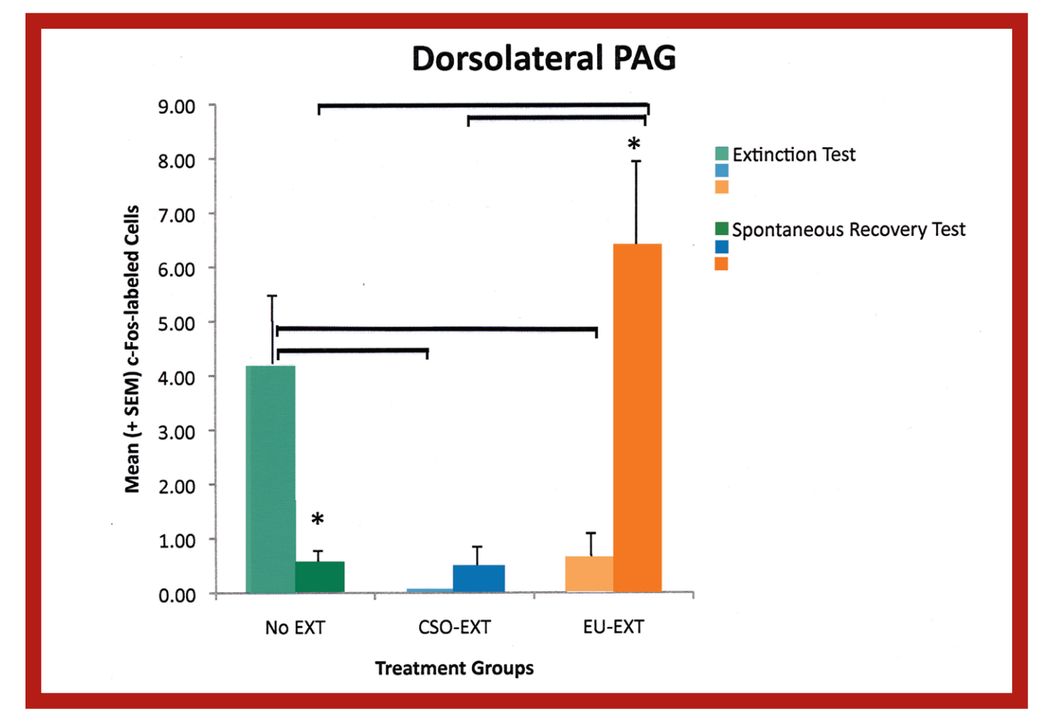
Dorsolateral PAG
Extinction Test
Spontaneous Recovery Test
Mean (+ SEM) c-Fos-labeled Cells
No EXT
CSO-EXT
EU-EXT
Treatment Groups

Figure 9:

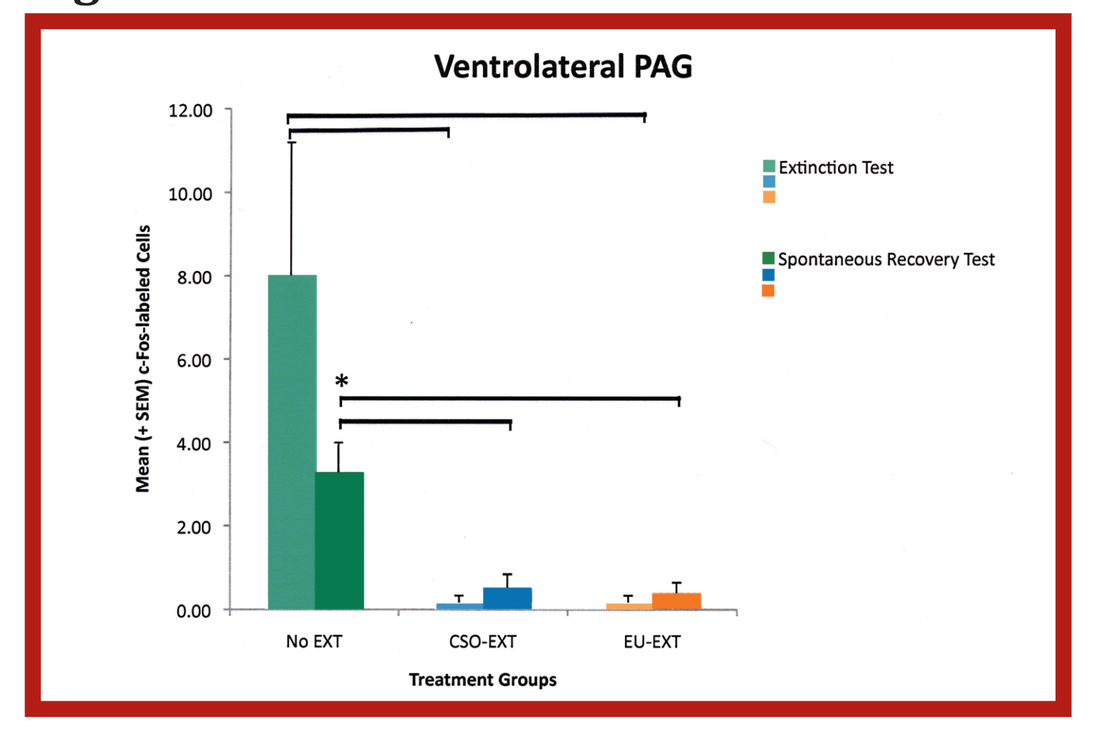
Ventrolateral PAG
Extinction Test
Spontaneous Recovery Test
Mean (+ SEM) c-Fos-labeled Cells
No EXT
CSO-EXT
EU-EXT
Treatment Groups

Table 2: Correlations between behavioral measures and *c-fos* expression in PAG's longitudinal columns. Enhanced SAC consumption at the SR test is positively correlated with increases in *c-fos* protein in dlPAG neurons.

Behavioral Measure	Dorsomedial PAG	Dorsolateral PAG	Lateral PAG	Ventrolateral PAG	Total PAG
Days to Asymptotic EXT (N=39)	-.31*	NS	-.32*	NS	-.32*
SAC Consumed at Asymptotic EXT (N=32)	-.64*	-.35*	-.70*	-.57*	-.75*
SAC Consumed at SR test (N=18)	NS	.56*	-.70*	-.72*	NS

* =p<0.05

Summary and Conclusions

- Extinction learning that employed the EU-EXT procedure of disassociating the CS and US produced more rapid extinction of a CTA and also inhibited spontaneous recovery of this defensive reaction to a learned fear.
- Immunohistochemical analyses indicated that there were no differences between the *c-fos* protein expression in mPFC, GNC or AMY of the rats that went through CSO-EXT versus EU-EXT.
- *C-fos* expression in PAG was significantly lower in rats that underwent either CTA extinction procedure (CSO or EU) as compared to animals that maintained a CTA (NE group).
- Our data indicate that the number of neurons expressing *c-fos* protein in the dlPAG is higher in rats failing to show a significant SR (EU-EXT group) as compared to those exhibiting a SR of a CTA (CSO group).
- These data are consistent with other studies indicating that the midbrain PAG, and the dlPAG in particular, are important in modulating defensive responses to conditioned fears (Bittencourt et al., 2004; Moreira et al., 2009).
- The number of cells expressing *c-fos* during SR in the PAG is small. But note that others have reported that very small populations (e.g., <1%) of neurons in other brain areas can produce dramatic behavioral changes (see Witten et al., 2010).
- Our data are correlative and do not indicate that more *c-fos* in the dlPAG causes suppression of SR.

Conclusion:

- *C-fos* protein expression in the PAG and the dlPAG, in particular, is enhanced in rats that underwent EU-EXT and do not show a SR of a CTA. Future studies are aimed at determining if manipulation of dlPAG can reverse or potentiate these effects thereby revealing the extent to which dlPAG controls SR of this defensive reaction to a learned fear.

References and Acknowledgements

Bittencourt, A.S., Carobrez, A.P., Zamprogno, L.P., Tufik, S & Schenberg, L.C. (2004) Organization of single components of defensive behaviors within distinct columns of Periaqueductal gray matter of the rat: Role of N-methyl-D-Aspartic acid glutamate receptors. *Neuroscience.* 125, 71-89.

Floyd, N.S., Price, J.L., Ferry, A.T., Keay, K.A. & Bandler, R. (2000) Orbitomedial prefrontal cortical projections to distinct longitudinal columns of te Periaqueductal gray in the rat. *The Journal of Comparative Neurology*. 422, 556-578.

Garcia J., Kimeldorf D.J., & Knelling R.A. (1955) Conditioned aversion to saccharin resulting from exposure to gamma radiation. *Science*, 122,157-158.

Hsu, S.M., Raine, L., & Fangler, H. (1981) Use of avidin-biotin peroxidase complex (ABC) in immunoperoxidase techniques: A comparison between ABC and unlabeled antibody (PAP) procedures. *Journal of Histochemistry & Cytochemistry, 29*, 577-580.

Lamprecht, R., & Dudai, Y. (1996) Transient expression of c-fos in rat amygdala during training is required for encoding conditioned taste aversion memory, *Learning & Memory, 3*, 31-41.

McNally G.P., Pigg M., & Weidemann G. (2004) Opioid receptors in the midbrain periaqueductal gray regulate extinction of pavlovian fear conditioning. *Journal of Neuroscience, 24,* 6912-6919.

McNally, G.P., Lee, B.-W., Chiem, J.Y. & Choi, E.A. (2005) The midbrain Periaqueductal gray and fear extinction: Opioid receptor subtype and roles of cyclic AMP, protein kinase A, and mitogen-activated protein kinase. *Behavioral Neuroscience, 119*, 1023-1033.

Mickley, G.A., Kenmuir, C.L., McMullen, C. A., Yocom, A. M., Valentine, E. L., Dengler-Crish, C.M., Weber, B., Wellman, J. A. & Remmers-Roeber, D. R. (2004) Dynamic Processing of Taste Aversion Extinction in the Brain. *Brain Research, 1061/1*, 79-89.

Mickley, G.A., Kenmuir, C.L., Yocom, A.M., Wellman, J.A., & Biada, J.M. (2005) A Role for prefrontal cortex in extinction of a conditioned taste aversion. *Brain Research, 1051*, 176-182.

Mickley, G.A., Hoxha, Z., Bacik, S., Kenmuir, C.L., Wellman, J.A., Biada, J.M. & DiSorbo, A. (2007). Spontaneous recovery of a conditioned taste aversion differentially alters extinction-induced changes in c-Fos protein expression in rat amygdala and neocortex. *Brain Research, 1152*, 139-157.

Mickley, G.A., Disorbo, A., Wilson, G.N., Huffman, J., Bacik, S., Hoxha, Z., Biada, J.M. & Kim, Y.-H. (2009) Explicit disassociation of a conditioned stimulus and unconditioned stimulus during extinction training reduces both time to asymptotic extinction and spontaneous recovery of a conditioned taste aversion. *Learning & Motivation, 40,* 209-220.

Moreira, F.A., Aguiar, D.C., Campos, A.C., Lisboa, S.F., Terzian, A.I., Resstel, L.B. & Guimaraes, F.S. (2009) Antiaversive effects of cannabinoids: Is the periaqueductal gray involved? *Neural Plasticity*, 2009. Retrieved from: http://www.ncbi.nlm.nih.gov/pmc/articles/PMC2593468/pdf/NP2009-625469.pdf

Nolan, L.J., McCaughey, S.A., Giza, B.K., Rhinehart-Doty, J.A., Smith, J.C., & Thomas, R.S. (1997) Extinction of a conditioned taste aversion in rats: I. Behavioral effects. *Physiology and Behavior, 61*, 319-323.

Pare, D., Quirk, G.J., & Ledoux, J.E. (2004) New vistas on amygdala networks in conditioned fear. *Journal of Neurophysiology*. 92, 1-9.

Parker L.A, 2003. Taste avoidance and taste aversion: evidence for two different processes. *Learning & Behavior, 31*, 165-172.

Paxinos, G. & Watson, C. (1998) *The rat brain in stereotaxic coordinates*. Academic Press, New York.

Peters, J., Kalivas, P.W. & Quirk, G.J. (2009) Extinction circuits for fear and addiction overlap in prefrontal cortex. *Learning & Memory*. 16, 279-288.

Price, J.L. (2005) Free will versus survival: brain systems that underlie intrinsic constraints on behavior. *Journal of Comparative Neurology*. 493, 132-139.

Thomas, B.L., Longo, C.L. & Ayres, J.J.B. (2005) Thwarting the renewal (relapse) of conditioned fear with the explicitly unpaired procedure: Possible interpretations and implications for treating human fears and phobias. *Learning and Motivation*, *36*, 374-407.

Ulrich-Lai, Y.M. & Herman, J.P. (2009) Neural regulation of endocrine and autonomic stress responses. *Nature Reviews, Neuroscience.* 10, 397-409.

Witten, I.B., Lin, S.-C., Brodsky, M., Prakash, R., Diester, I., Anikeeva, P., Gradinaru, V., Ramakrishnan, C. & Deisseroth, K. (2010) Cholinergic interneurons control local circuit activity and cocaine conditioning. *Science. 330*, 1677-1681.

Acknowledgements:

The authors wish to acknowledge the following students and technicians for their contributions to this work: Doug Placko, Ashley Sandmann, James Romanchik, Sarah Clark, Jennifer Dunger, Sarah Frischmann, Jennifer Graebert, Nick Grisak, Natalie Hogan, Bruce Kinley, Ye-Hyun Kim, & Dave Revta.

Supported by NIMH grant: 2-R15-MH063720-03.